98

The Institute of Cancer Research

CANCER RESEARCH UK

Discovery and validation of biomarkers to support clinical development of NXP800: A first-in-class orally active, small-molecule HSF1 pathway inhibitor

Paul A Clarke, Robert te Poele, Marissa Powers, Gary Box, Emmanuel de Billy, Alexis de Haven Brandon, Albert Hallworth, Angela Hayes, Henry McCann, Swee Sharp, Melanie Valenti, Florence I Raynaud, Suzanne A Eccles, Matthew Cheeseman, Keith Jones, Paul Workman

HSF1 and the oncogene-associated stress response

- HSF1 is an ancient transcription factor regulating the classical eukaryotic Heat Shock Response (HSR) and is a master regulator of proteotoxic stress[1]
- In response to oncogenic stress, HSF1 is hijacked in cancer cells to regulate a gene set overlapping with but distinct from the canonical HSR[2]
- Depleting HSF1 inhibits the oncogenic effects of p53 loss and RAS activation in mouse models[3] – indicating therapeutic potential
- HSF1 is amplified, over-expressed and activated in human cancers and these and the oncogenic stress signature predict clinical outcome in numerous human cancers, including ovarian[2,4] – indicating possible patient populations
- Preclinical toxicity indicates a potential therapeutic index - a phase 1 dose escalation, clinical study is ongoing

Identifying ARID1A as a predictive biomarker for NXP800

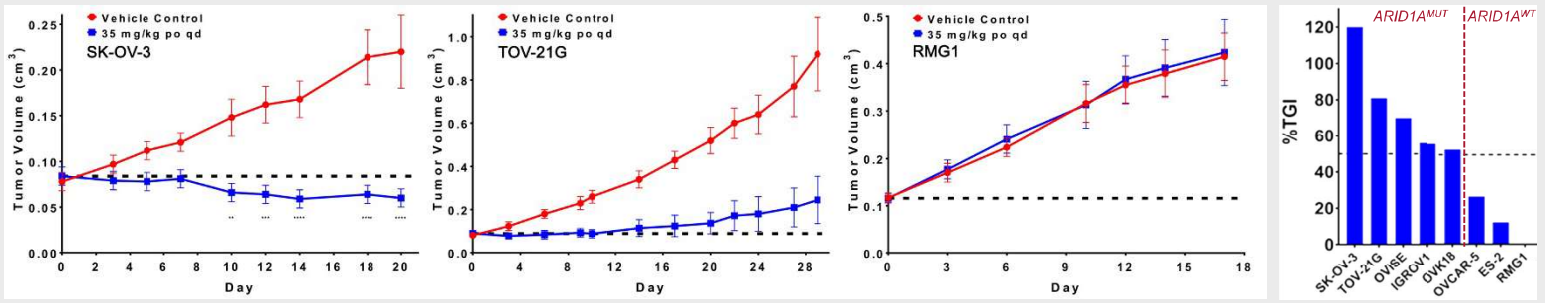
Vehicle Control
35 mg/kg po qd
SK-OV-3
TOV-21G
RMG1
Tumor Volume (cm³)
Day
%TGI
ARID1A^MUT
ARID1A^WT
SK-OV-3
TOV-21G
OVISE
IGROV1
OVK18
OVCAR-5
ES-2
RMG1

- NXP800 has activity in multiple xenograft models of ovarian clear cell cancer
 - all responsive ovarian cancer tumours xenografts (5 of 8; >50% Tumour growth inhibition) carried loss of function *ARID1A* mutations
 - ARID!A is a component of the large chromatin remodelling SWI/SNF complex that regulates transcription by altering their surrounding chromatin structure
 - ARID1A confers gene specificity to the SNF/SWI complex, particularly required for activation of genes normally repressed by surrounding chromatin structure
 - Impact of *ARID1A* mutation confirmed in isogenic models – ongoing; data not shown

Sanger Institute cancer cell line panel

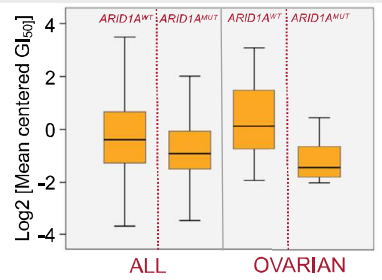
Log2 [Mean centered GI50]
ARID1A^WT
ARID1A^MUT
ALL
OVARIAN

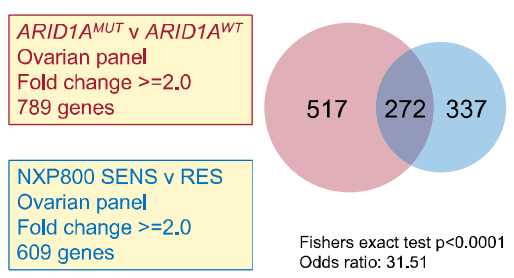
ARID1A^MUT v ARID1A^WT
Ovarian panel
Fold change >=2.0
789 genes
NXP800 SENS v RES
Ovarian panel
Fold change >=2.0
609 genes
517
272
337
Fishers exact test p<0.0001
Odds ratio: 31.51

- An *ARID1A* mutation was a predictive biomarker for NXP800 sensitivity (4.7-fold; p_{adj}<0.001) in the ovarian cancer cell line subset
 - *ARID1A* mutation status not statistically predictive for the whole panel
 - possibly predictive for gastric and intestinal cancers
 - ovarian subset: 36% of basal genes associated with NXP800-sensitivity are also ARID1A-reg[d]

Discovery and profile of NXP800

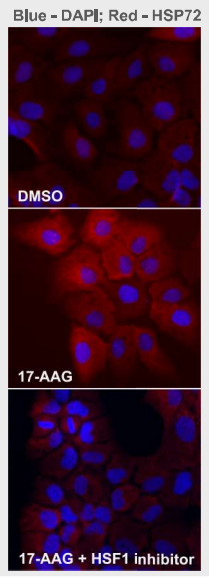
Blue - DAPI; Red - HSP72
DMSO
17-AAG
17-AAG + HSF1 inhibitor

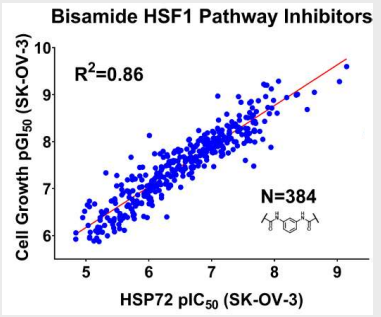
Bisamide HSF1 Pathway Inhibitors
R²=0.86
N=384
Cell Growth pGI50 (SK-OV-3)
HSP72 pIC50 (SK-OV-3)

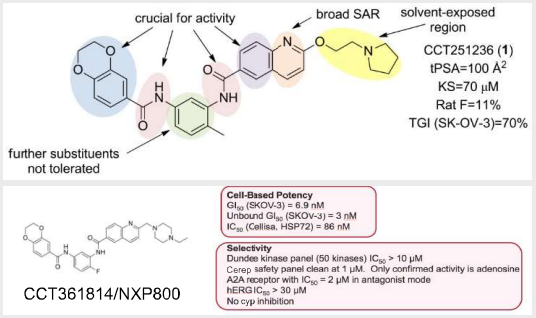
crucial for activity
broad SAR
solvent-exposed region
further substituents not tolerated
CCT251236 (1)
tPSA=100 Å²
KS=70 μM
Rat F=11%
TGI (SK-OV-3)=70%
CCT361814/NXP800
Cell-Based Potency
Selectivity

- HSF1 pathway inhibitors identified by the ability to block the induction of HSP72 by HSP90 inhibitor 17-AAG
 - screened 200K compounds from the AstraZeneca collection
 - discovered *N,N'*-4-methyl-1,3-phenylenediamide core structure
 - screen run in U2OS human osteosarcoma cells, followed up in SK-OV-3 human ovarian cancer cells
 - highly potent, low nanomolar hit against the HSF1 pathway and cancer cell growth inhibition
 - optimised through lead compound/chemical tool to clinical candidate[5,6]
 - NXP800 has a clean profile across *in vitro* hERG, Cyp450, kinase and Cerep-Safety-87 assays

Identifying protein and gene signature biomarkers for PD with NXP800

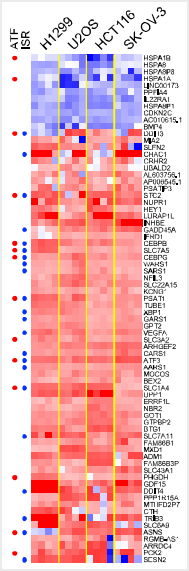
ATF
ISR
H1299
U2OS
HCT116
SK-OV-3

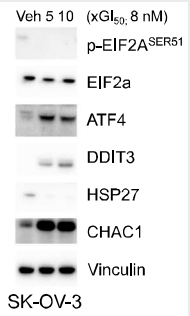
Veh 5 10 (xGI50, 8 nM)
p-EIF2A^SER51
EIF2a
ATF4
DDIT3
HSP27
CHAC1
Vinculin
SK-OV-3

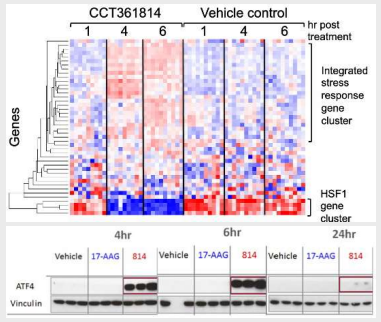
CCT361814
Vehicle control
hr post treatment
Genes
Integrated stress response gene cluster
HSF1 gene cluster
4hr
6hr
24hr
Vehicle
17-AAG
814
ATF4
Vinculin

- Multiple cancer cell types treated with 5x GI_{50} NXP800 for 6 h and RNAseq profiled
 - significant enrichment of elevated integrated stress response (ISR) and ATF4-regulated genes
 - immunoblotting confirmed induction of ISR and ATF4
- NanoString probeset confirmed elevated ISR/ATF4 and repressed HSF1 activity *in vivo*
 - immunoblotting confirmed induction of ATF4 protein

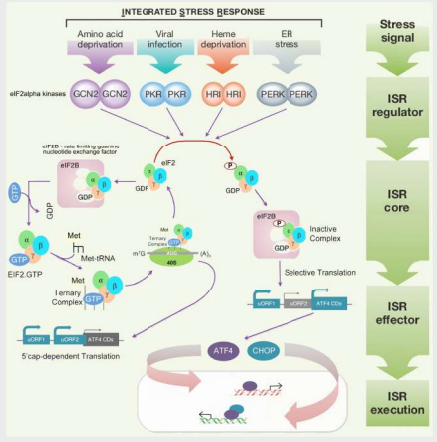
INTEGRATED STRESS RESPONSE
Amino acid deprivation
Viral infection
Heme deprivation
ER stress
Stress signal
ISR regulator
ISR core
ISR effector
ISR execution
GCN2
PKR
HRI
PERK
ATF4
CHOP
Inactive Complex
Selective Translation
5'cap-dependent Translation

- NXP800 treatment induces prolonged activation of the ISR[7]
 - ISR activation blocks HSF1 activation
 - prolonged ISR/ATF4 activation and CHOP induction can result in cell death

Phase 1 clinical trial of NXP800

- Phase 1 trial comprising a dose-escalation (Phase 1a) and expansion (Phase 1b) phases commenced and first patient treated in January 2022 (www.clinicaltrials.gov/ct2/show/NCT05226507)
 - in the Phase 1b, the safety and preliminary anti-tumor activity of NXP800 will be evaluated in ARID1A biomarker-selected patients – initially those with ovarian clear cell carcinoma and endometrioid carcinoma

Summary and future plans

- NXP800 is the first-in-class, orally active inhibitor of HSF1 activation that is important for cancer cells
- Biomarkers discovered and validated to provide PK/PD relationship and Pharmacologic Audit Trail
 - strong validation for modulating HSF1 activation in a range of human cancers
 - biomarkers indicate a mechanism of action involving activation of the integrated stress response
- Initial clinical studies will focus on ovarian clear cell carcinoma and endometrioid carcinoma – with a very high unmet need
 - cancers with *ARID1A* loss show increased sensitivity to NXP800, providing a patient biomarker
 - therapeutic potential in additional tumour types
- Multiple orthogonal approaches are being applied to identify the precise key molecular target(s)

Contact

Dr. Paul Clarke or Prof. Paul Workman
Centre for Cancer Drug Discovery, The Institute of Cancer Research
15 Cotswold Road, Sutton, SM2 5NG, United Kingdom
Email: Paul.Clarke@icr.ac.uk, Workman@icr.ac.uk

Acknowledgments

We acknowledge funding from CRUK, Battle Against Cancer Investment Trust (BACIT) and Sixth Element Capital. NXP800 is licenced to Nuvectis Pharma which is finding its development.

References

1. Li J et al. 2017. Trends Cell Biol. 27:895.
2. Mendillo ML et al. 2012. Cell 150:549.
3. Dai C et al. 2007. Cell 130:1005.
4. Powell CD et al. 2016. PLoS 11:e168389.
5. Cheeseman MD et al. 2017. J. Med. Chem. 60:180.
6. Rye CS et al. 2016. Med. Chem. Comm. 2016 7:1580.
7. Pakos-Zebrucka K et al. 2016. EMBO Rep. 17:1374.

SIXTH ELEMENT CAPITAL LLP

NuvectisPharma, Inc.